AACR Abstract Number: **3869**

Proteomics highlights which G-protein coupled receptors are candidates for ADC development

Juergen Kast[1], Robert Boyd[1], James Ackroyd[1], Jason Allen[1], Amanda Anderson[1], Martin Barnes[1], Ludmila Bozhenok[1], Palminder Dusanjh[1], Lindsey Hudson[1], Xiaohong Yu[1], Dee Aud[2], Rahel Awdew[2], Arnima Bisht[2], Deepal Pandya[2], Michael Trang[2], Jonathan Terrett[2], Christian Rohlff[1]

[1]Oxford BioTherapeutics Ltd, Abingdon, United Kingdom; [2]Oxford BioTherapeutics Inc, San Jose, CA

Introduction

Antibody-drug conjugates (ADCs) are a recent and exciting development for targeted therapy of cancer. Their efficacy is governed by ADC-intrinsic characteristics such as avidity, drug load and linker chemistry, and mechanisms of activation and action, which can be controlled or clarified in the early stages of ADC development. In contrast, the properties that define a promising ADC target are still somewhat unclear. One key aspect is the abundance of a target on cancer cells across various cancer types relative to various critical normal tissues. While mRNA profiling is limited in this respect, quantitative proteomics of plasma membranes is ideally suited for a rapid assessment of cancer specify and abundance.

A recent bioinformatics prediction, the protein family of G-protein coupled receptors (GPCRs) encompasses 899 distinct members in the human genome. These cell surface receptors are the target of more than one third of conventional drugs, yet their potential for ADCs is largely unexplored. Due to its non-targeted nature, proteomics can identify and quantify thousands of proteins per sample and generate maps that can be used to select the GPCRs that hold the most promise to become ADC targets.

OGAP is a unique proteomic database that integrates information at the tissue, disease and protein isoform level across diseases, indications, and normal tissues to clarify protein expression levels and profiles. Built over the course of more than a decade, and recently focusing exclusively on oncology, it currently holds information on ~4,000,000 human protein peptide sequences, ~16,000 human proteins sequenced, ~7,000 cancer membrane proteins, ~50 tissues/organs, and ~60 diseases,

Building on OGAP and a proprietary sample preparation and processing workflow that relies on state-of-the-art high-throughput mass spectrometry and data processing to provide quantitative information on over 4,000 membrane-enriched proteins per tissue sample we have established a novel predictive tool to establish each protein's potential to serve as a target for ADC development.

Using the above approach we have identified 255 GPCRs. The expression of these GPCRs in normal and cancer tissues was assessed using the protein index, a composite score of two factors: prevalence (in %) x abundance (in %), Its maximum value is 10000 (100% x 100%). Differential expression criteria were then applied to these GPCRs (> 2 fold T/N, > 5 fold T/N and Tumor expression only). 112 cancer specific GPCRs were identified as candidate targets for follow up studies.
For a cancer antigen to have therapeutic application it should have low or absent expression in key organs. In contrast high expression in a disease-related and/or non-vital normal tissues does not necessary disqualify a GPCR candidate.

Figure 1. Comparison of GPCR protein index and mRNA profiles for 5 GPCRs.

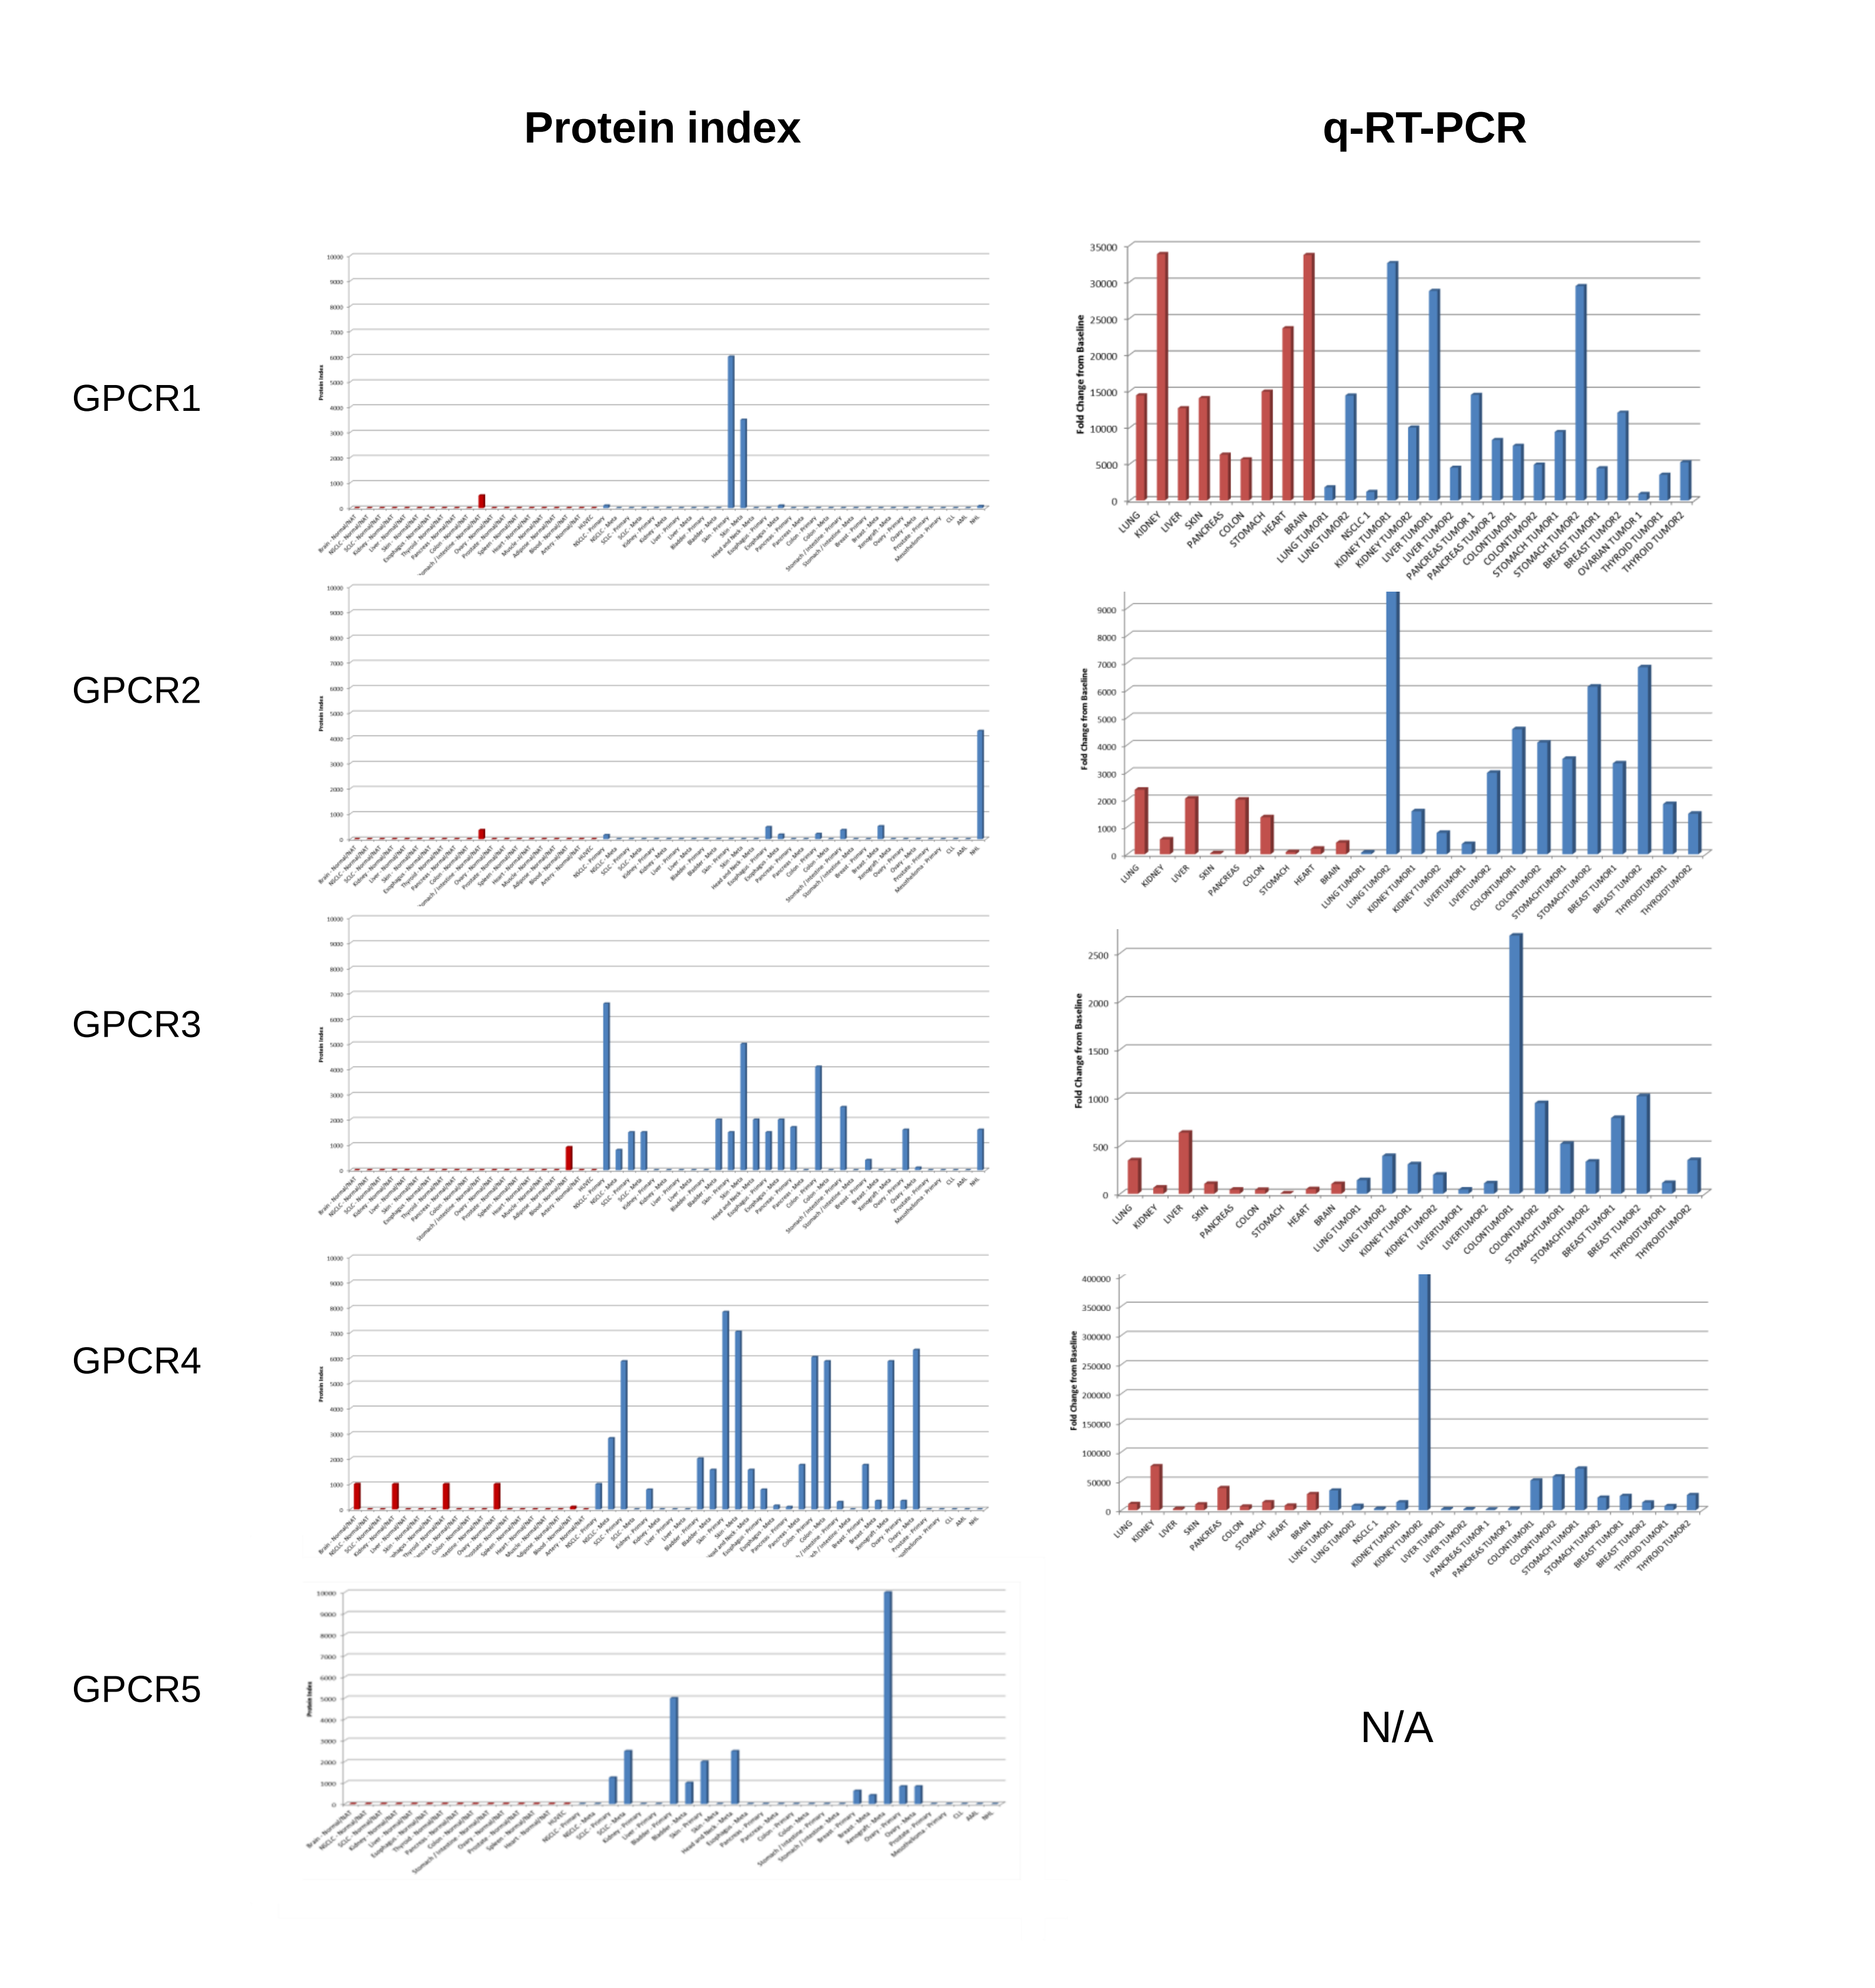
Protein index
q-RT-PCR
GPCR1
GPCR2
GPCR3
GPCR4
GPCR5
N/A

Normal tissue expression of protein or mRNA is shown by red bars. Cancer tissue expression of protein or mRNA is shown by blue bars.

Figure 2. Normal tissue expression is a key determinant in GPCR candidate target selection.

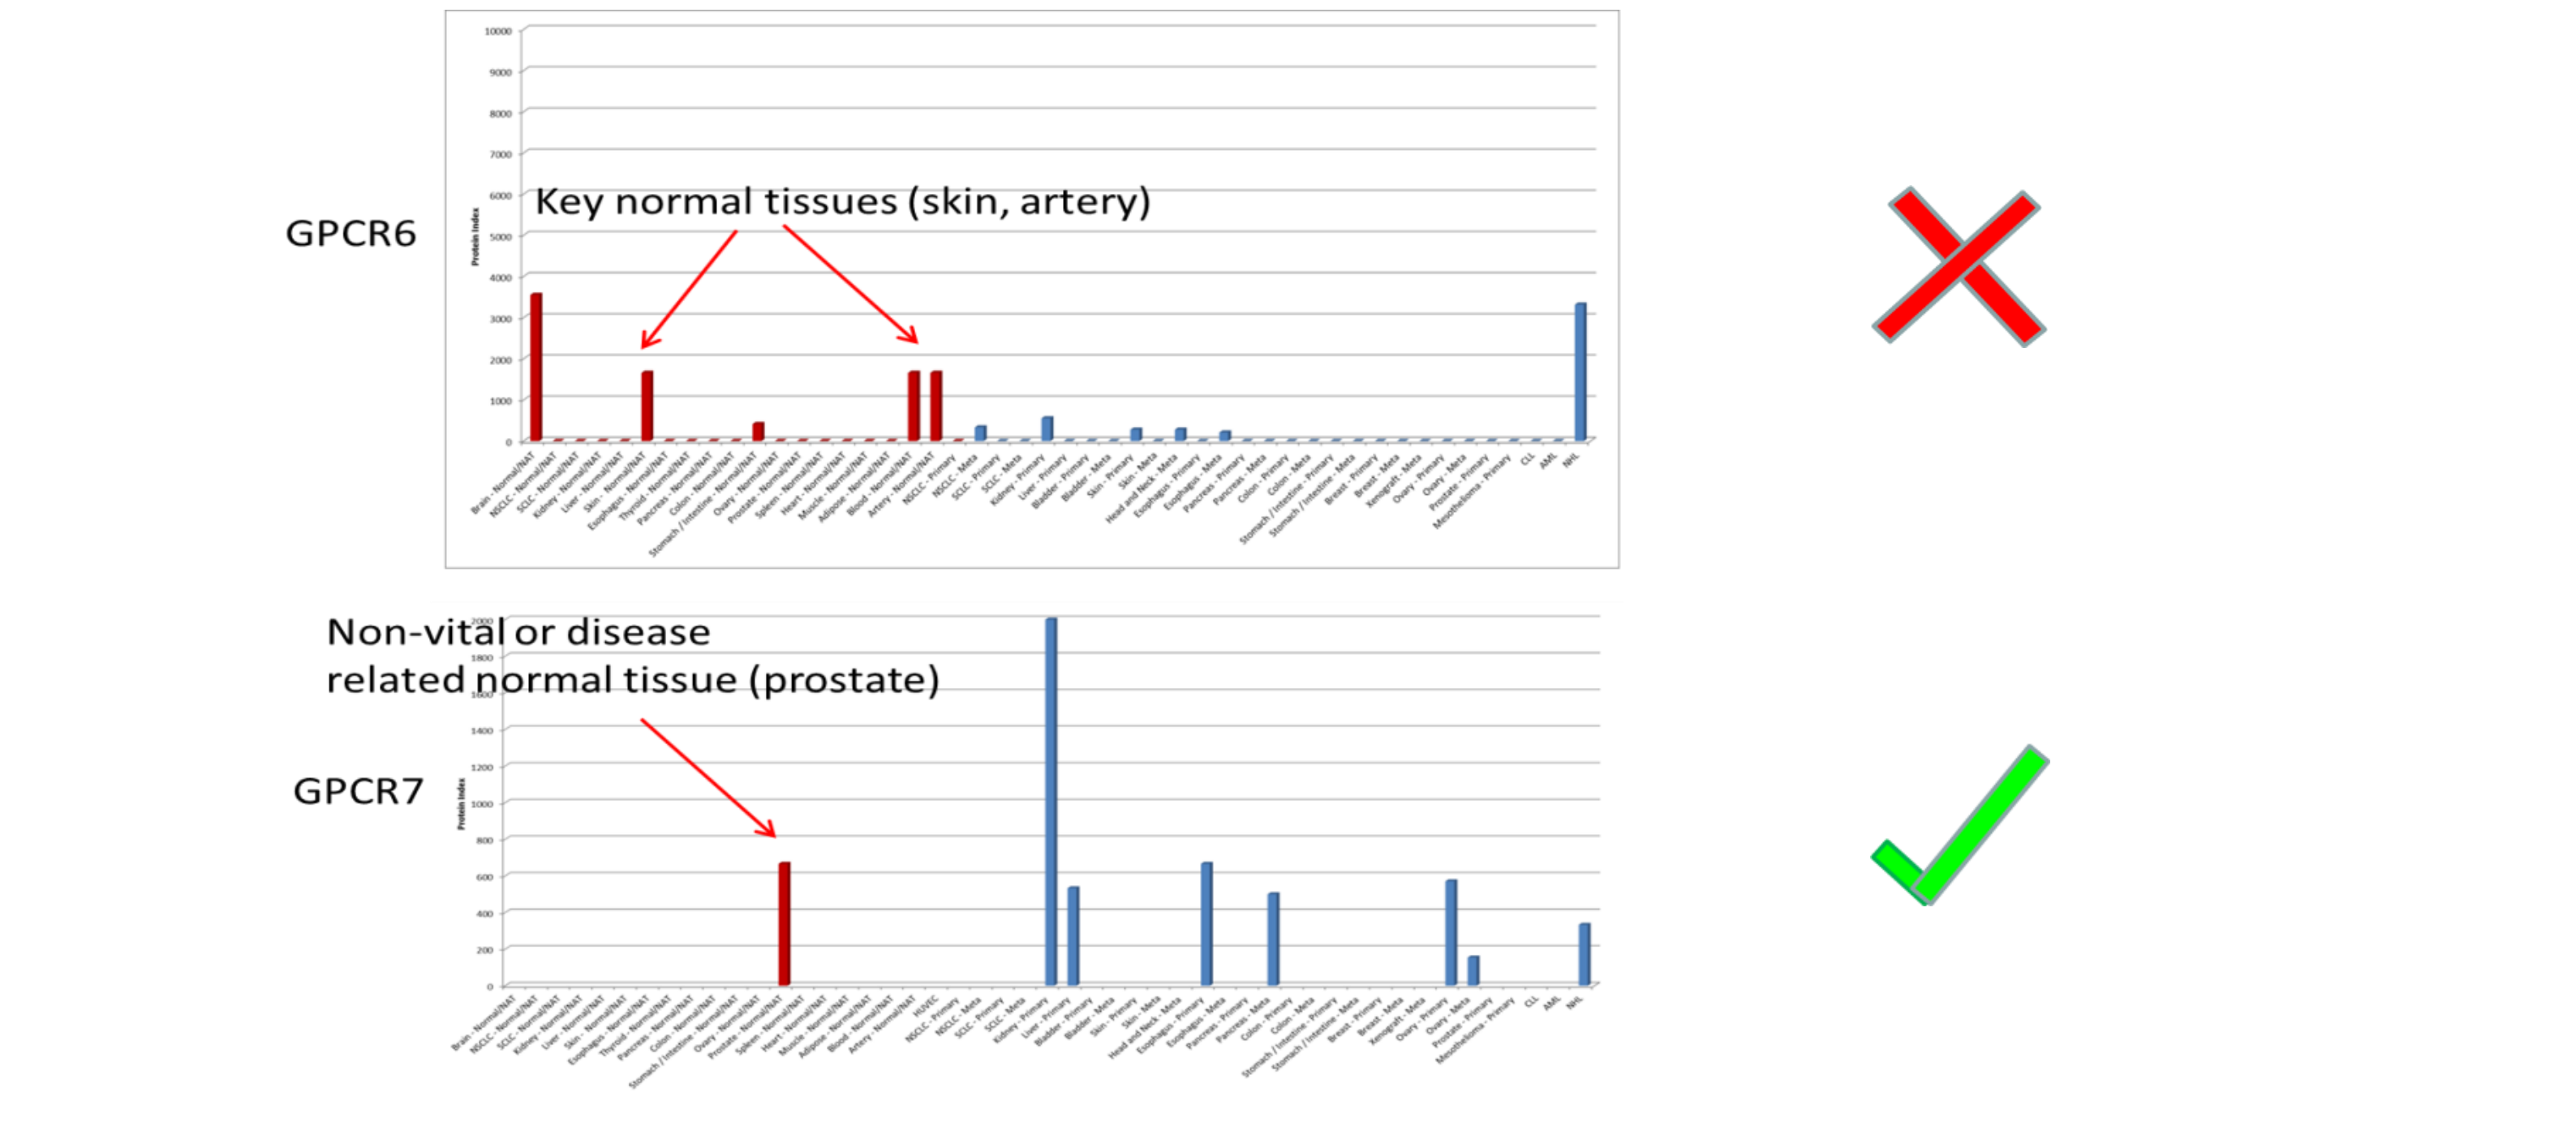
GPCR6
Key normal tissues (skin, artery)
Non-vital or disease
related normal tissue (prostate)
GPCR7

Figure 3. On the basis of further validation studies (A) mAbs were generated against a GPCR target on a cell line (B).

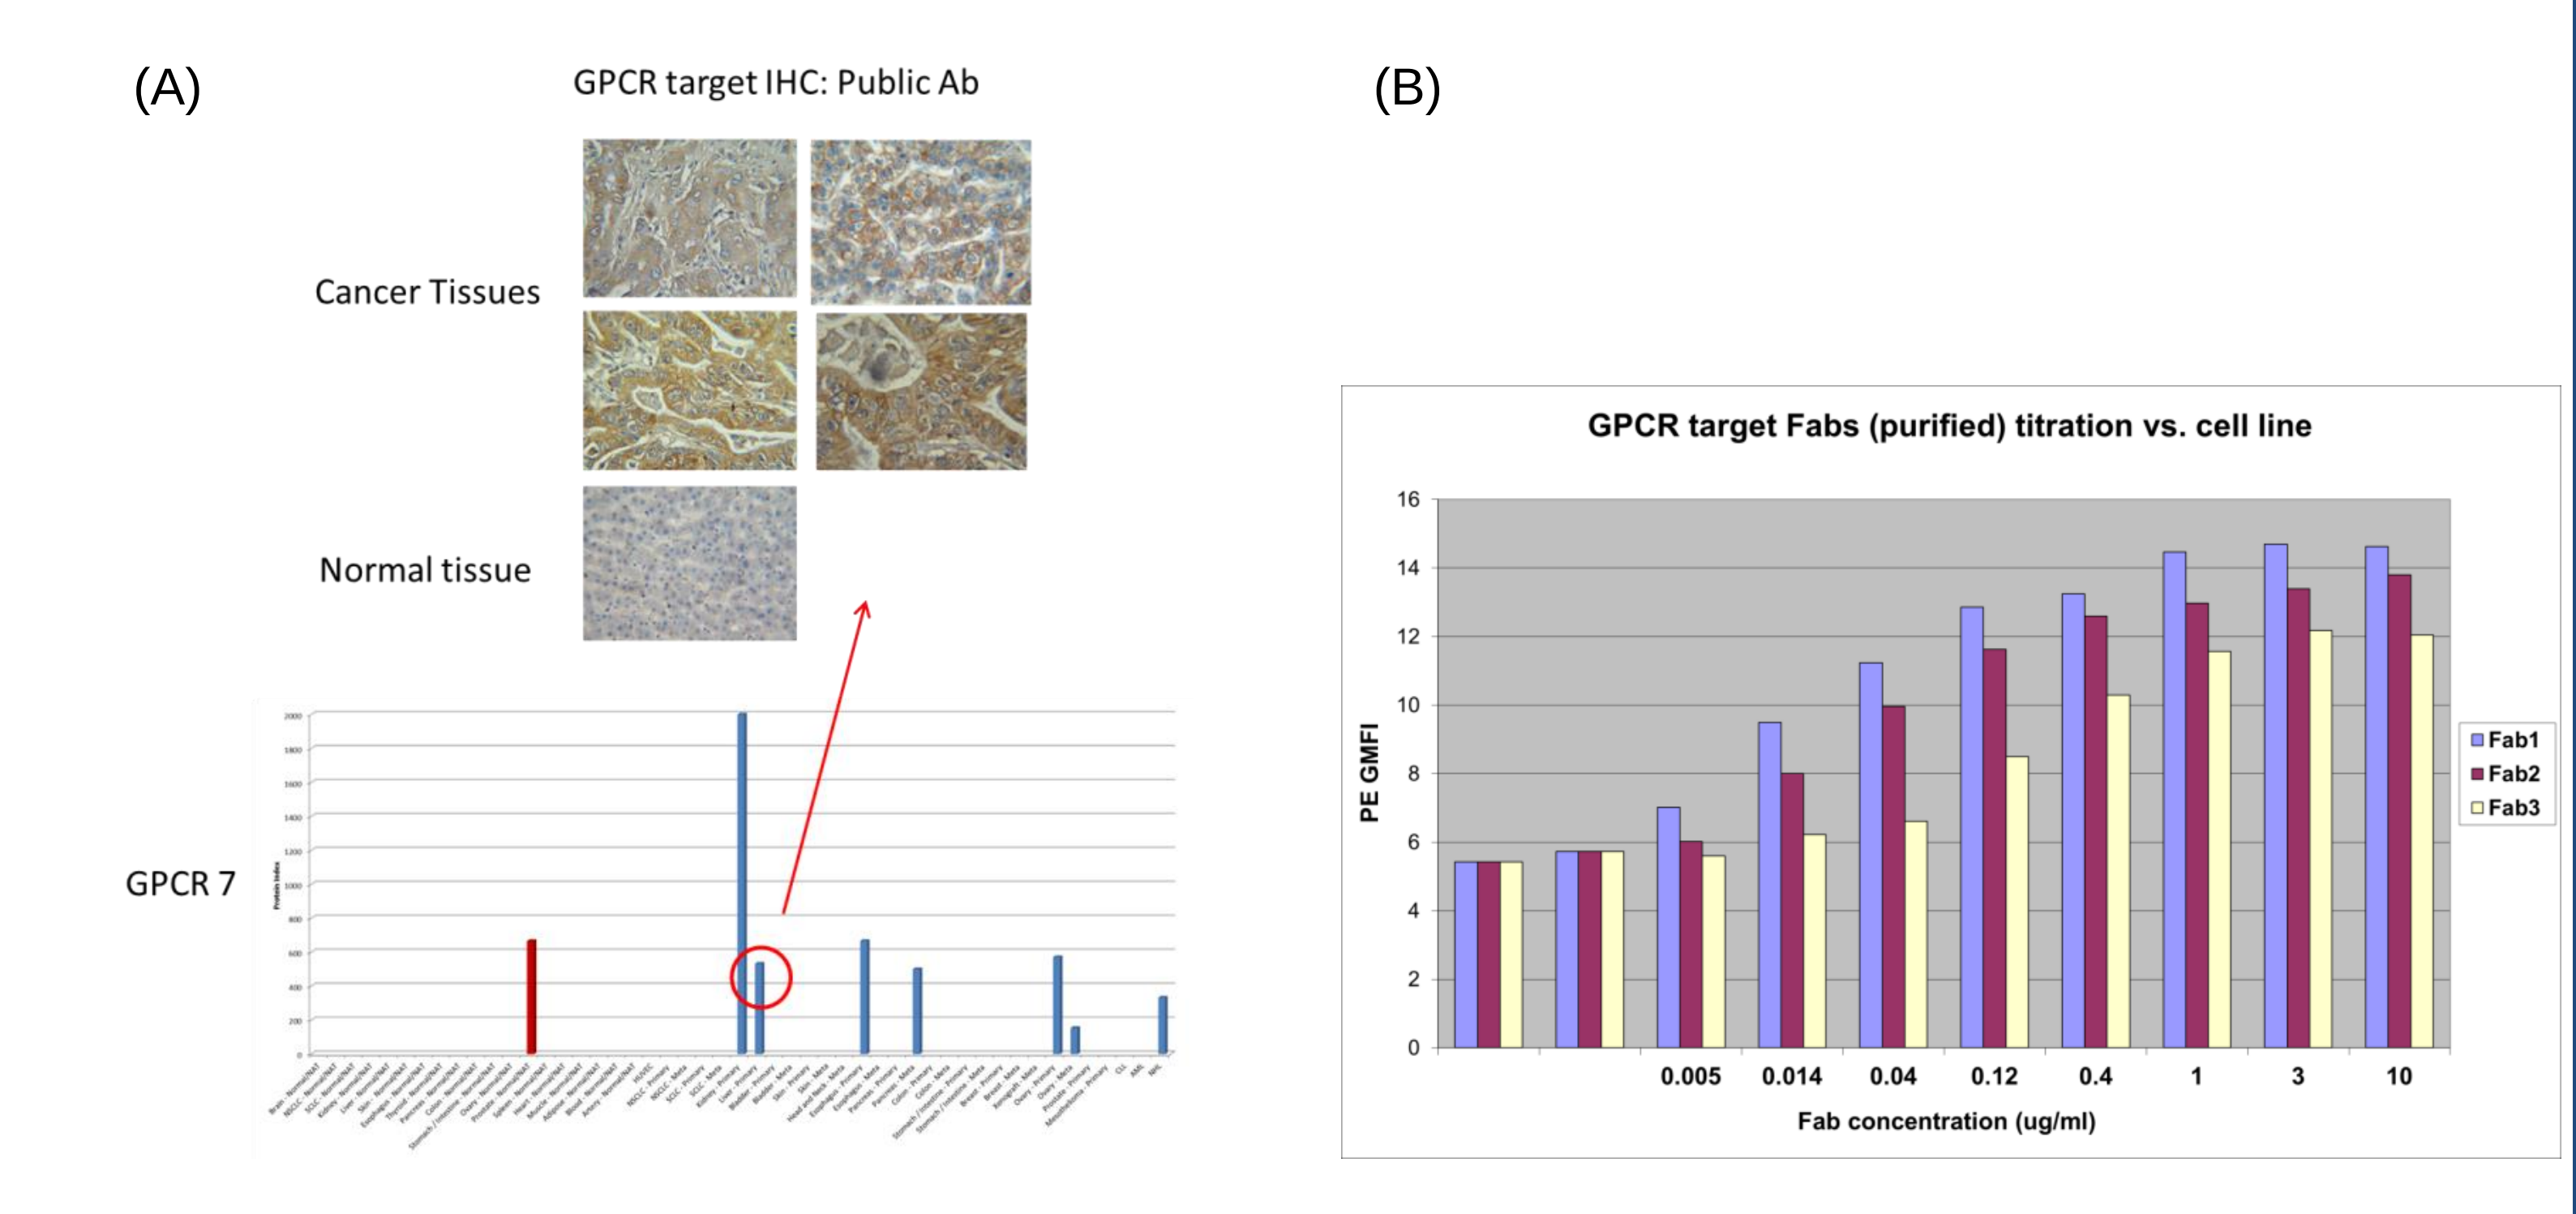
(A)
GPCR target IHC: Public Ab
Cancer Tissues
Normal tissue
GPCR 7
(B)
GPCR target Fabs (purified) titration vs. cell line
PE GMFI
Fab1
Fab2
Fab3
0.005
0.014
0.04
0.12
0.4
1
3
10
Fab concentration (ug/ml)

Conclusions

- The OGAP database has been used successfully to identify GPCRs that are candidates for ADC development.
- Information obtained by using a high-throughput quantitative proteomics approach has been critical for this assessment.
- In contrast, mRNA profiles obtained for these GPCRs did not prove useful for the selection of ADC targets and priority indications in these cases.
- While demonstrated here for GPCRs, this approach can be used for any family of membrane proteins, or indeed the entire membrane proteome.